An Open-Label Trial Assessing Short- and Long-Term Tolerability and Efficacy of ZYN002 (Cannabidiol) Administered as a Transdermal Gel to Children and Adolescents with 22q11.2 Deletion Syndrome (INSPIRE)

Helen Heussler,[1] Jonathan Cohen,[2] Caroline Buchanan,[3] Carol O'Neill,[4] Stephen O'Quinn,[4] Terri Sebree[4]

[1]Centre for Clinical Trials in Rare Neurodevelopmental Disorders, Children's Health Queensland Hospital and Health Services, South Brisbane, Queensland, AU; [2]Genetic Clinics Australia, North Caulfield, Victoria, AU; [3]Greenwood Genetic Center, Greenville, SC, USA; [4]Zynerba Pharmaceuticals, Devon, PA, USA

BACKGROUND: 22q11.2 DELETION SYNDROME

- 22q11.2 deletion syndrome (22q) is the second most common chromosomal disorder after Down syndrome[1]
- 22q occurs in 1 in 3000 to 1 in 6000 live births[2]
- The deletion of genes such as *TBX1* may be responsible for characteristic signs (such as heart defects, a cleft palate, distinctive facial features, hearing loss, and low calcium levels) and behavioral problems
- Children with 22q are at increased risk for several psychiatric disorders, including anxiety, social withdrawal, attention-deficit hyperactivity disorder (ADHD), cognitive impairment, autism spectrum disorder (ASD), and mood disorders, as well as psychotic disorders and schizophrenia (adolescence/adulthood)[3]
- Approximately 40% of these children suffer from some type of anxiety disorder[4]
 - Specific phobias are most common, followed by generalized anxiety disorder, separation anxiety, and obsessive-compulsive disorder[5]
- Early negative experience has been shown to increase the risk of anxiety and is associated with atypical development of the physiological stress response[6]
 - Serious medical complications, such as congenital heart disease, feeding difficulties, and multiple surgeries or hospitalizations, may also predispose some children to anxiety disorders due to stressors associated with repeated medical procedures or poor sense of control over one's body[7]
- The presence of anxiety symptoms in children with 22q is negatively correlated with adaptive function and impacts everyday living skills[8]

BACKGROUND: CANNABIDIOL AND ZYN002

- Disruption in the endocannabinoid system (ECS) is one of the proposed mechanisms underlying symptoms affecting children with neurodevelopmental disorders[9]
- Cannabidiol acts as a negative allosteric modulator at presynaptic type 1 cannabinoid receptors[10]
- Cannabidiol has also shown activity at serotonin $5HT1_A$,11 gamma-aminobutyric acid A,[12] and dopamine D2 and D3[13,14] receptors
- ZYN002 (also known as Zygel™) is a pharmaceutically produced (not plant-derived) cannabidiol transdermal gel in clinical development for the treatment of behavioral symptoms associated with 22q and Fragile X syndrome (FXS)
- ZYN002 does not contain delta-9-tetrahydrocannabinol (THC) and avoids conversion to THC in the stomach due to transdermal application[15]

WHY STUDY ZYN002 IN 22q?

- Anxiety and a constellation of behavioral symptoms are similar between 22q, FXS, and ASD[16]
- ZYN002 has been shown to have potential benefit in FXS[17] and ASD[18]
- Dysfunction in the ECS may be present in all 3 disorders[9]
- ZYN002 does not contain THC. THC may increase the risk for schizophrenia,[19] and THC-containing cannabinoid products should be avoided in individuals with 22q since they are at increased risk of psychosis

Figure 1. INSPIRE Phase 2 Trial Design

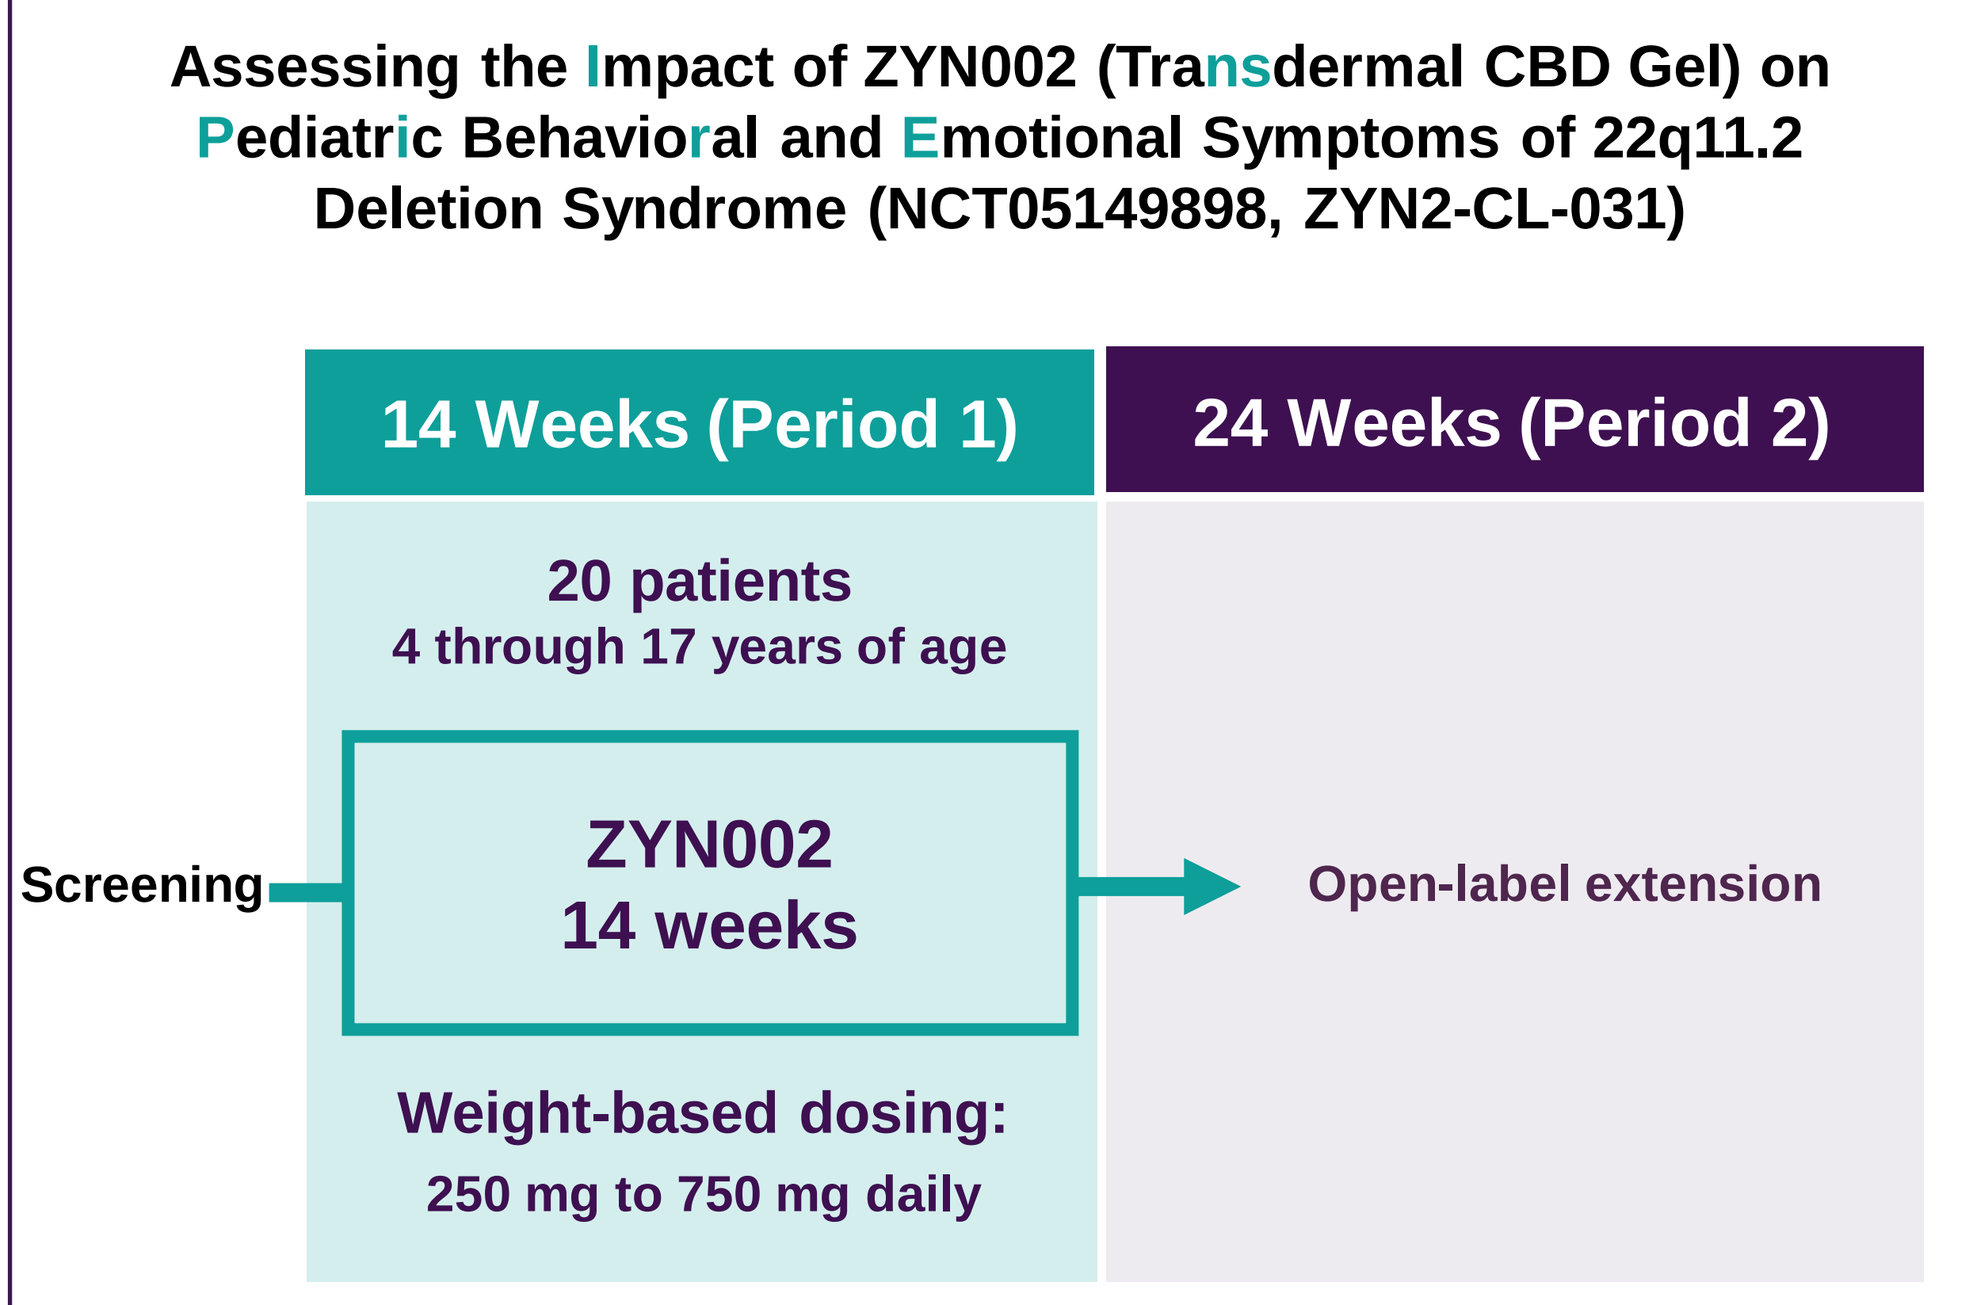
Assessing the Impact of ZYN002 (Transdermal CBD Gel) on Pediatric Behavioral and Emotional Symptoms of 22q11.2 Deletion Syndrome (NCT05149898, ZYN2-CL-031)
14 Weeks (Period 1)
24 Weeks (Period 2)
20 patients
4 through 17 years of age
Screening
ZYN002
14 weeks
Open-label extension
Weight-based dosing:
250 mg to 750 mg daily

METHODS

- 3 sites: 2 in Australia and 1 in the United States
- Children and adolescents ages 4 through 17 with genetically confirmed 22q who also have the following:
 - Pediatric Anxiety Rating Scale-Revised (PARS-R) score of ≥10 at Screening and Visit 2 (Day 1)
 - Clinical Global Impression-Severity (CGI-S) score of ≥4 at Screening and Visit 2 (Day 1)
- Primary outcome measure: Safety and tolerability
- Secondary outcome measures to assess efficacy:
 - PARS-R
 - Anxiety, Depression, and Mood Scale (ADAMS)
 - Aberrant Behavior Checklist-Community (ABC-C)
 - Clinical Global Impression-Improvement (CGI-I)
 - Qualitative Caregiver Reported Behavioral Problems Survey
- Patients received ZYN002 250 mg (weight ≤35 kg) or 500 mg (weight >35 kg) daily, in divided doses every 12 hours, as an add-on to standard-of-care medications
- At Week 6, if the patient had less than a 25% improvement from baseline in the ABC-C irritability subscale, the investigator could increase the dose as follows:
 - Patients who weighed ≤35 kg receiving a total daily dose of 250 mg ZYN002 could increase the dose to 500 mg
 - Patients who weighed >35 kg receiving a total daily dose of 500 mg ZYN002 could increase the dose to 750 mg/day
- Patients with a ≥35% improvement in the ABC-C irritability subscale were allowed to continue treatment for an additional 24 weeks (Period 2)

RESULTS

- 20 patients were enrolled; 17 completed 14 weeks **(Period 1)**; reasons for the 3 discontinuations included:
 - Adverse event unrelated to ZYN002, n=1
 - Patient withdrew consent, n=1
 - Lost to follow-up, n=1
- Mean age 9.9 years (range 4-15 years); 60% were male
- 53% (8/15 patients assessed) had ASD by Autism Diagnostic Observation Schedule-2 (ADOS®-2)
- One patient's dose increased from 250 to 500 mg/day at week 6 and 2 at week 14; 1 patient increased from 500 mg to 750 mg/day at week 14
- At the end of **Period 1** (Week 14) and **Period 2** (Week 38), statistically significant improvements were seen in all scales of the ADAMS, ABC-C, and PARS-R (Figures 2-4)
- 13 patients entered **Period 2**; 12 completed 38 weeks, with 1 patient discontinuing due to withdrawn consent

- The PARS-R is a clinical interview that assesses symptoms of anxiety and generates a composite score for:
 - Overall frequency of anxiety symptoms
 - Overall severity of anxiety feelings
 - Overall avoidance of anxiety-provoking situations
 - Interference with family relationships and/or performance at home
 - Interference with peer and adult relationships and/or performance outside of home
- At Baseline, patients had **clinically significant moderate anxiety** on average (PARS-R=15) **(Figure 2)**
- At Weeks 14 and 38, patients had improved to **minimal to mild anxiety** on average (PARS-R=5-10) **(Figure 2)**

Figure 2. Significant Improvements in Anxiety as Measured by the PARS-R

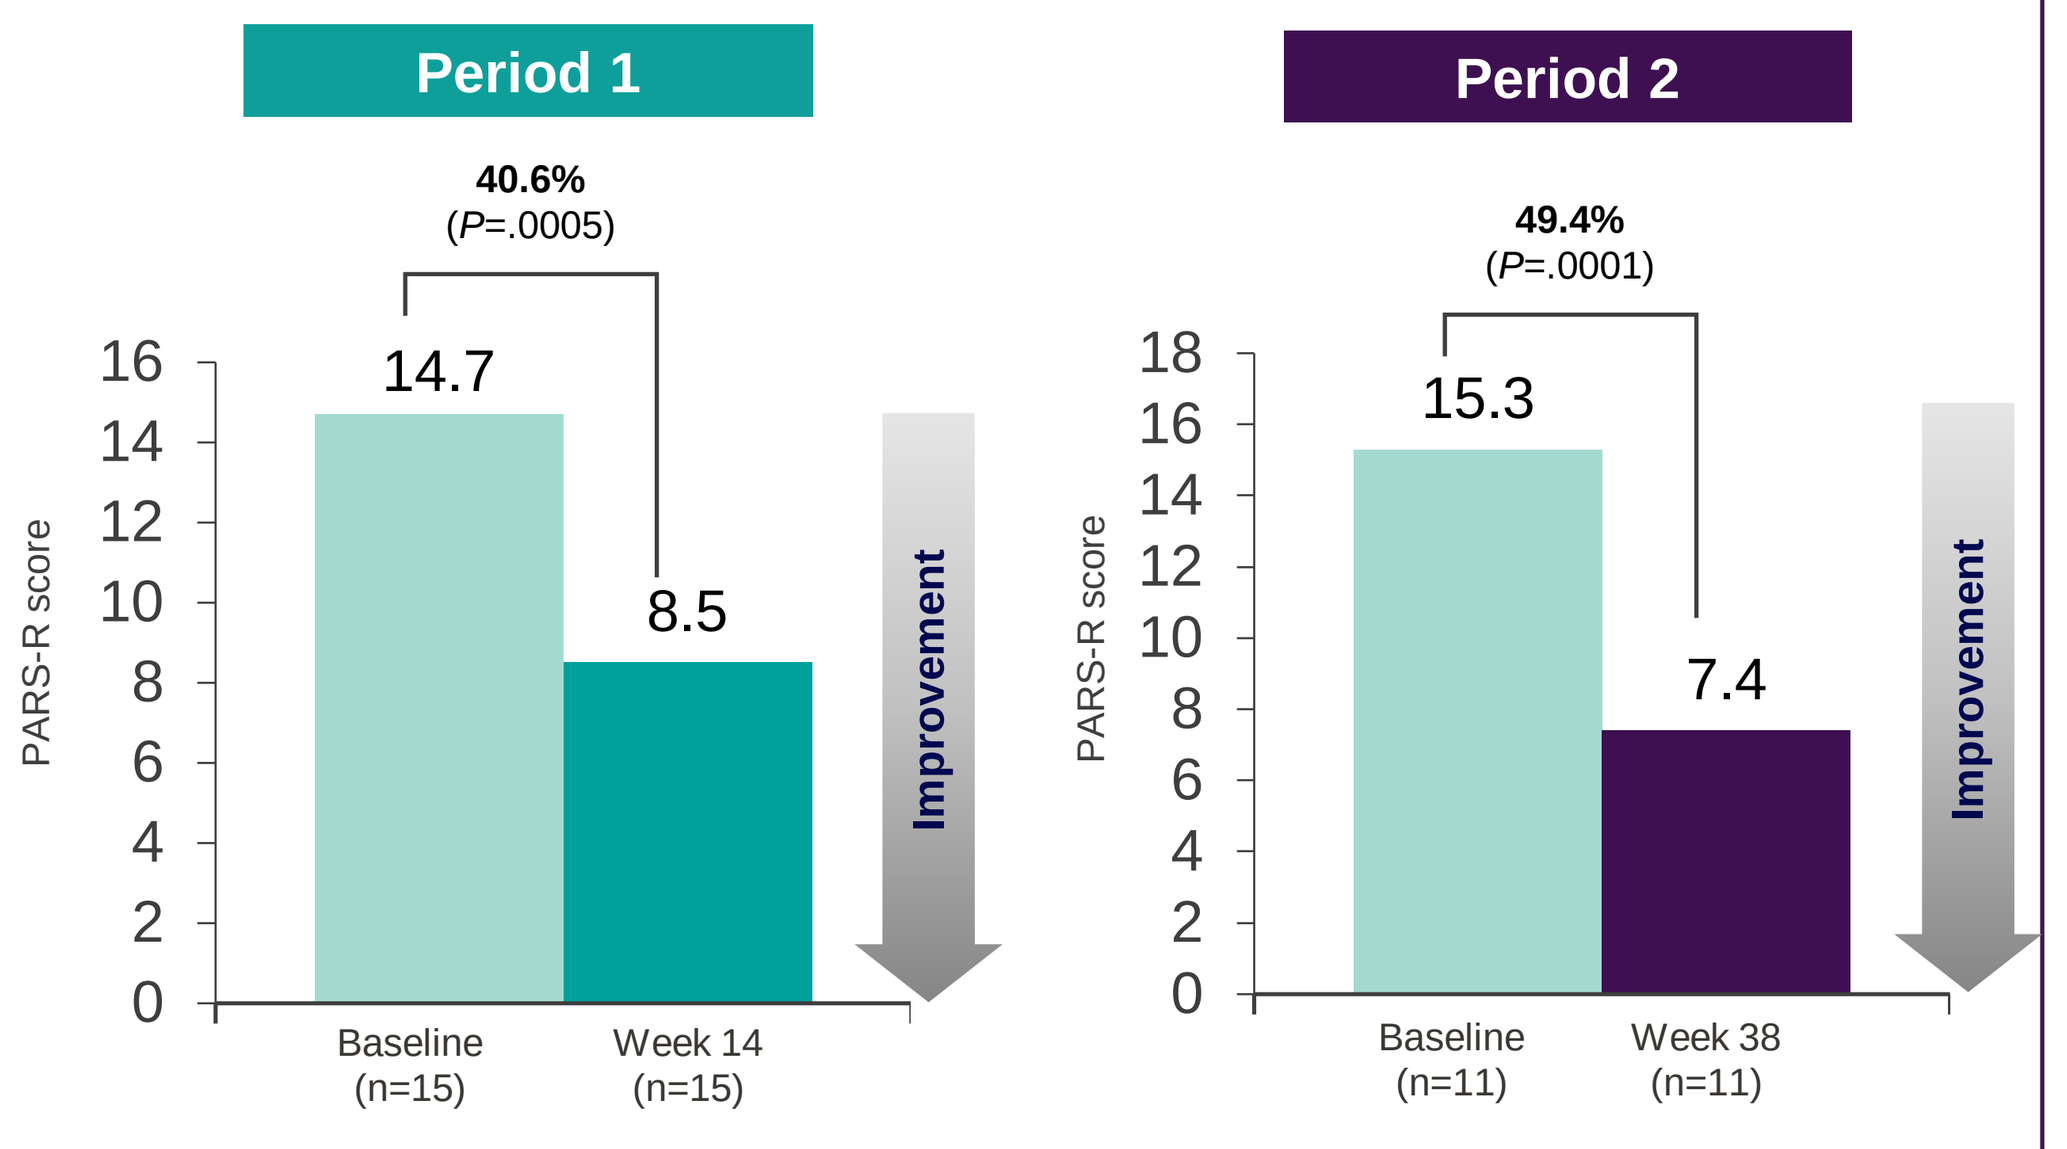
Period 1
40.6%
(P=.0005)
PARS-R score
14.7
8.5
Baseline (n=15)
Week 14 (n=15)
Improvement
Period 2
49.4%
(P=.0001)
PARS-R score
15.3
7.4
Baseline (n=11)
Week 38 (n=11)
Improvement

The efficacy analysis for Period 1 included 15 patients for PARS-R (2 patients did not have valid assessments at Week 14).
The efficacy analysis for Period 2 included 11 patients; 1 patient who completed Week 38 stopped drug prior to completion.

Clinically Meaningful Improvements in CGI-I (Period 1)

- 75% of patients were rated by clinicians as "improved," "much improved," or "very much improved"
- 62.5% of patients were rated by clinicians as "much improved" or "very much improved"

Table 1. Adverse Events and Safety Over 38 Weeks

	Patients (n=20) % Patients (n patients or events)
Treatment-Emergent Adverse Events (TEAEs)[a]	65% (13 patients; 30 events)
Mild in severity	86.7%
Treatment-Related AEs	15% (3 patients; 3 events)
Application site pain Application site itching *All were mild and transient*	10% (2 patients; 2 events) 5% (1 patient; 1 event)
Discontinuations due to TEAEs unrelated to ZYN002	1 patient
Serious AEs All unrelated to ZYN002	3 (3 patients)
No clinically significant changes in laboratory, vital signs, or electrocardiograms	

[a]Treatment-emergent adverse events are any events, whether related or unrelated to ZYN002.

Figure 3. Significant Improvements in ADAMS Total and All Subscale Scores

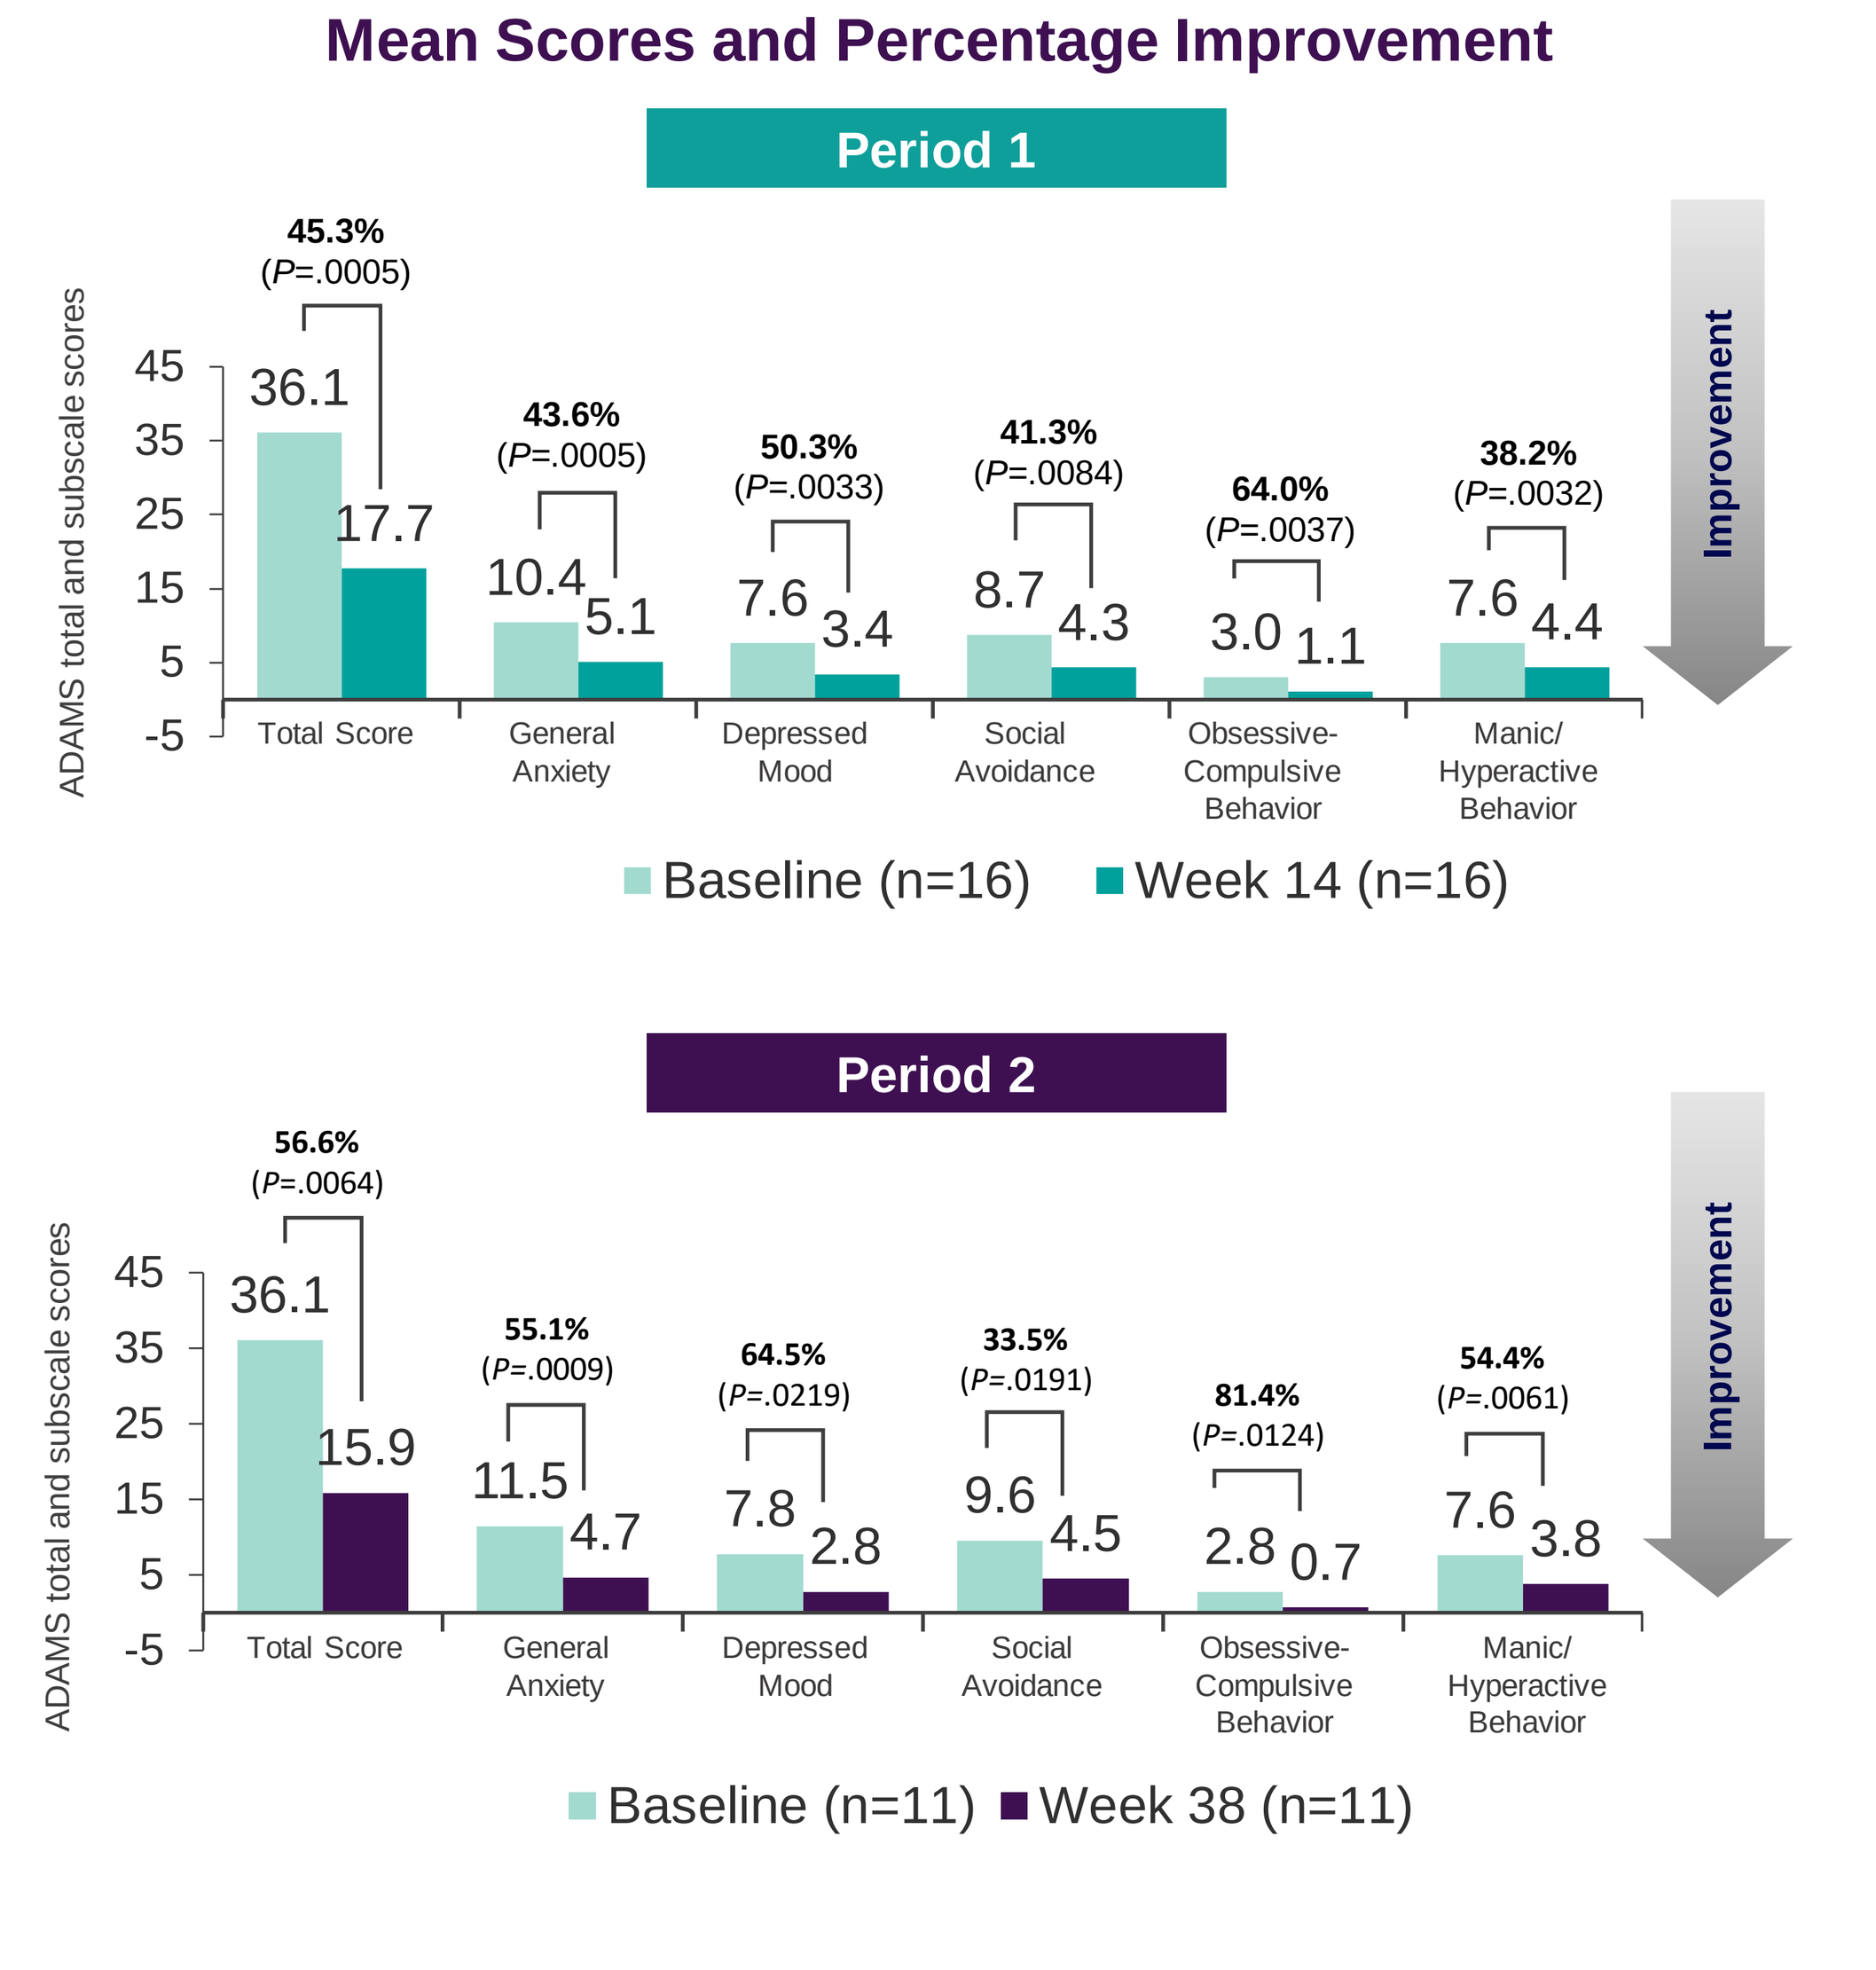
Mean Scores and Percentage Improvement
Period 1
ADAMS total and subscale scores
Total Score: 36.1, 17.7; 45.3% (P=.0005)
General Anxiety: 10.4, 5.1; 43.6% (P=.0005)
Depressed Mood: 7.6, 3.4; 50.3% (P=.0033)
Social Avoidance: 8.7, 4.3; 41.3% (P=.0084)
Obsessive-Compulsive Behavior: 3.0, 1.1; 64.0% (P=.0037)
Manic/Hyperactive Behavior: 7.6, 4.4; 38.2% (P=.0032)
Baseline (n=16)
Week 14 (n=16)
Improvement
Period 2
ADAMS total and subscale scores
Total Score: 36.1, 15.9; 56.6% (P=.0064)
General Anxiety: 11.5, 4.7; 55.1% (P=.0009)
Depressed Mood: 7.8, 2.8; 64.5% (P=.0219)
Social Avoidance: 9.6, 4.5; 33.5% (P=.0191)
Obsessive-Compulsive Behavior: 2.8, 0.7; 81.4% (P=.0124)
Manic/Hyperactive Behavior: 7.6, 3.8; 54.4% (P=.0061)
Baseline (n=11)
Week 38 (n=11)
Improvement

The efficacy analysis for Period 1 included 16 patients for ADAMS (1 patient did not have a valid assessment at Week 14).
The efficacy analysis for Period 2 included 11 patients; 1 patient who completed Week 38 stopped drug prior to completion.
Total Score and Obsessive/Compulsive Behavior N=10 as the score for item #16 was missing at Week 38.

Figure 4. Significant Improvements in All ABC-C Subscale Scores

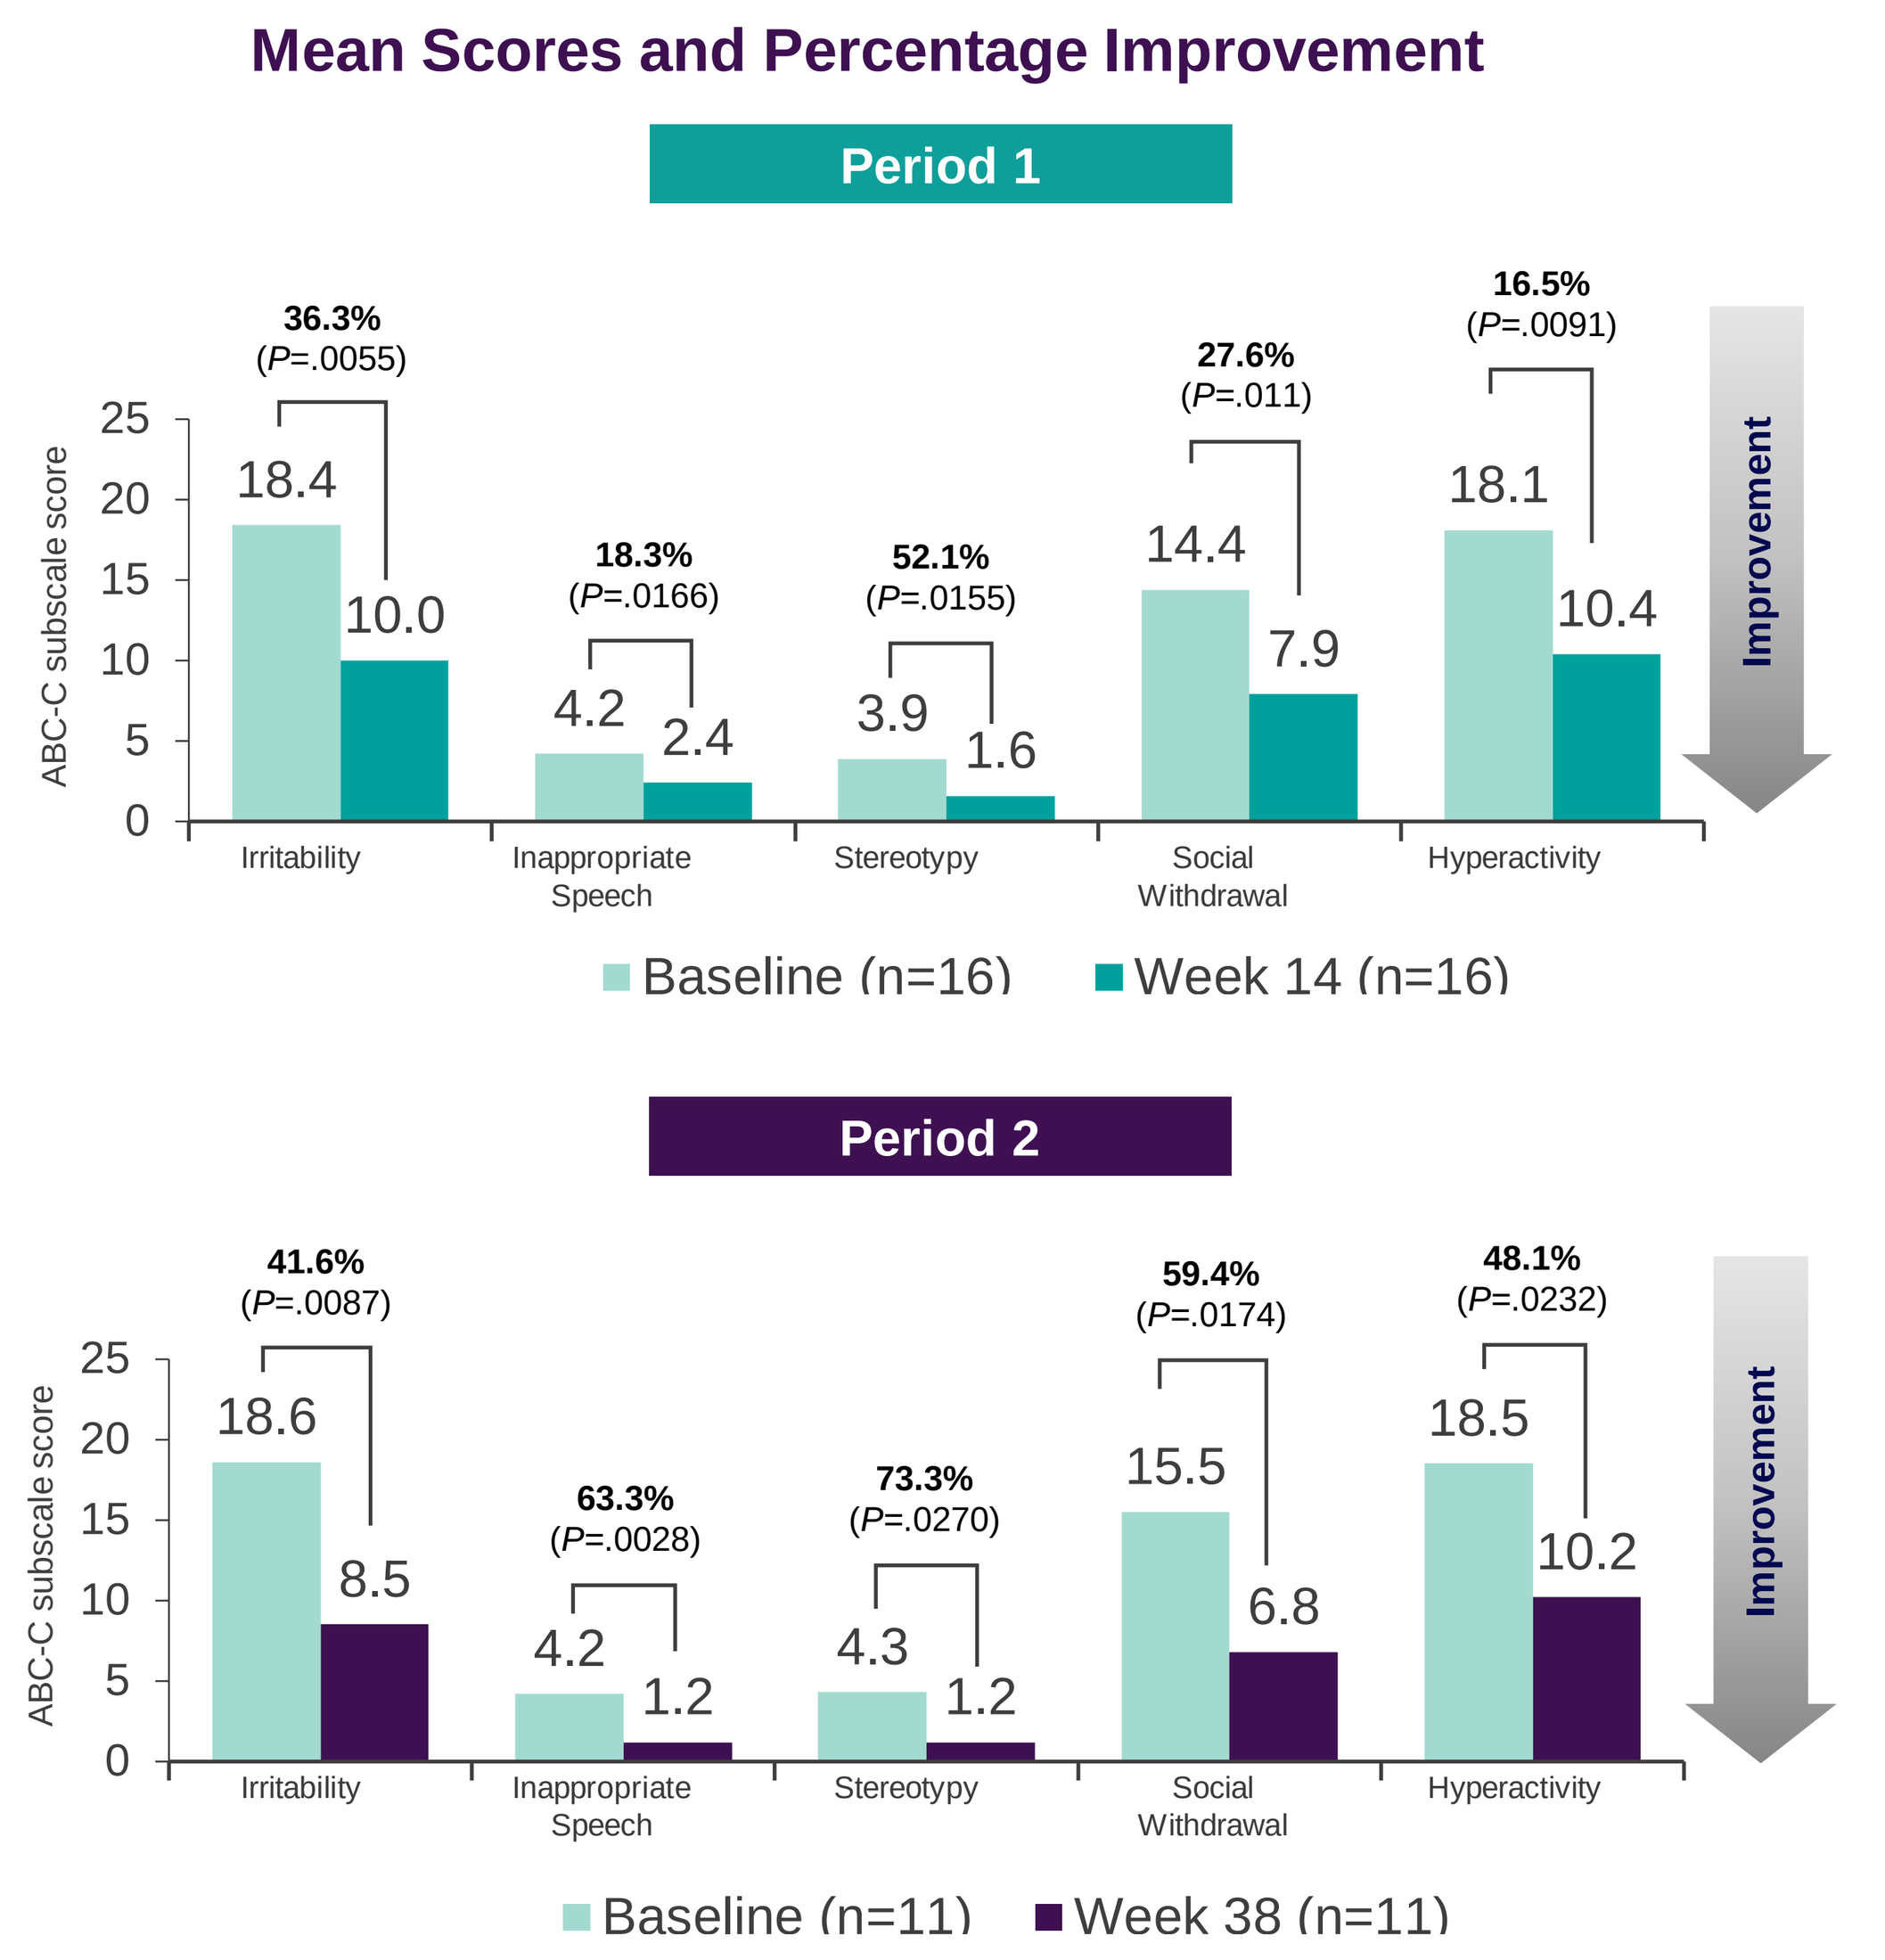
Mean Scores and Percentage Improvement
Period 1
ABC-C subscale score
Irritability: 18.4, 10.0; 36.3% (P=.0055)
Inappropriate Speech: 4.2, 2.4; 18.3% (P=.0166)
Stereotypy: 3.9, 1.6; 52.1% (P=.0155)
Social Withdrawal: 14.4, 7.9; 27.6% (P=.011)
Hyperactivity: 18.1, 10.4; 16.5% (P=.0091)
Baseline (n=16)
Week 14 (n=16)
Improvement
Period 2
ABC-C subscale score
Irritability: 18.6, 8.5; 41.6% (P=.0087)
Inappropriate Speech: 4.2, 1.2; 63.3% (P=.0028)
Stereotypy: 4.3, 1.2; 73.3% (P=.0270)
Social Withdrawal: 15.5, 6.8; 59.4% (P=.0174)
Hyperactivity: 18.5, 10.2; 48.1% (P=.0232)
Baseline (n=11)
Week 38 (n=11)
Improvement

The efficacy analysis for Period 1 included 16 patients for ABC-C (1 patient did not have a valid assessment at Week 14).
The efficacy analysis for Period 2 included 11 patients. 1 patient who completed Week 38 stopped drug prior to completion.

CONCLUSIONS

- INSPIRE provides initial evidence suggesting a positive risk-benefit profile for ZYN002 in improving anxiety-related and behavioral symptoms in children and adolescents with 22q when added to a stable standard of care
- Statistically significant improvements reported in:
 - PARS-R
 - Total score and all 5 subscales of ADAMS
 - All 5 subscales of ABC-C
- Clinically meaningful improvements reported in CGI-I
- ZYN002 was well tolerated with a safety profile consistent with other ZYN002 clinical trials
- These findings warrant further study of ZYN002 in children and adolescents with 22q

REFERENCES

1. Bassett AS, et al. *J Pediatr.* 2011;159(2):332-9.e1.
2. McDonald-McGinn DM, et al. *Nat Rev Dis Primers.* 2015;1:15071.
3. Swillen A, McDonald-McGinn DM. *Am J Med Genet C Semin Med Genet.* 2015;169(2):172-181.
4. Jolin EM, et al. *J Affect Disord*. 2012;136(3):222-228.
5. Feinstein C, et al. *Biol Psychiatry*. 2002;51(4):312-318.
6. Elzinga BM, et al. *Psychoneuroendocrinology*. 2008;33:227-237.
7. Beaton EA, Simon TJ. *J Neurodev Disord*. 2011;3(1):68-75.
8. Angkustsiri K, et al. *J Dev Behav Pediatr*. 2012; 33(9):713-720.
9. Cheung KAK, et al. *Front Psychiatry.* 2021;12:643442.
10. Laprairie RB, et al. *Br J Pharmacol*. 2015;172(20):4790-4805.
11. Russo EB, et al. *Neurochem Res*. 2005;30(8):1037-1043.
12. Bakas T, et al. *Pharmacol Res*. 2017;119:358-370.
13. McGuire P, et al. *Am J Psychiatry*. 2018;175(3):225-231.
14. Bian YM, et al. *Acta Pharmacol Sin*. 2019;40(3):374-386.
15. Merrick J, et al. *Cannabis Cannabinoid Res.* 2016;1:102-112.
16. Heussler H, et al. 22nd SSBP International Research Symposium, September 4-6, 2019.
17. Heussler H, et al. *J Neurodev Disord.* 2019;11(1):16.
18. Heussler H, et al. 23rd SSBP International Research Symposium, September 9-10, 2021.
19. Vaucher J, et al. *Mol Psychiatry.* 2018;23;1287-1292.

ACKNOWLEDGEMENTS / DISCLOSURES

Acknowledgements

Editorial/medical writing support under the guidance of the authors was provided by *p*-value communications, and was funded by Zynerba Pharmaceuticals, Devon, PA, USA.

Disclosures

This presentation is intended to communicate scientific and medical information about data from clinical trials involving ZYN002. ZYN002 is an investigational treatment. This means that it is not approved for commercial distribution by government regulatory bodies, including the United States Food and Drug Administration (FDA). This presentation is not designed or intended to promote the use of ZYN002 or any other Company product in order to impact prescribing.

HH, JC, and CB have received research support from Zynerba Pharmaceuticals. CON, SOQ, and TS are employees of Zynerba Pharmaceuticals.

The trial was funded by Zynerba Pharmaceuticals.

Scan here to view a PDF of this poster. Copies obtained through quick response (QR) code are for personal use only and may not be reproduced without written permission from the authors.

Presented at the American Society of Clinical Psychopharmacology (ASCP) Annual Meeting | May 30-June 2, 2023 | Miami Beach, FL