Transdermal Cannabidiol (CBD) Gel for the Treatment of Focal Epilepsy in Adults

JOHN MESSENHEIMER, MD[1]; TERENCE O'BRIEN, MD, FRACP[2]; SAMUEL F. BERKOVIC, MD, FRACP[3]; JACQUELINE FRENCH, MD[4]; MARCEL O. BONN-MILLER, PHD[5]; DONNA GUTTERMAN, PHARMD[5]

1 Moncure, NC, USA; 2 Royal Melbourne Hospital, The University of Melbourne, Parkville, Vic, AU; 3 Epilepsy Research Centre, Heidelberg, Vic, AU; 4 NYU Langone School of Medicine, New York, NY, USA; 5 Zynerba Pharmaceuticals, Inc. Devon, PA, USA

INTRODUCTION

- Cannabidiol (CBD) is the primary noneuphoric cannabinoid found in cannabis[1]
- Evidence suggests that CBD can reduce seizures in patients with epilepsy[2]

OBJECTIVE

- Evaluate the safety and efficacy of ZYN002 (transdermal cannabidiol [CBD] gel) as adjunctive therapy for the treatment of adult focal seizures

METHODS

Study Conduct

- STAR 1 was a phase 2A, randomized, double-blind, placebo-controlled study of ZYN002 administered BID for 12 weeks to adults with focal seizures; patients were initially randomized 1:1:1 to 195 mg or 390 mg CBD daily, or placebo
- STAR 2 is the open-label extension study for completers of STAR 1
- All STAR 2 patients started on 390 mg CBD daily with the option to increase the dose to 585 mg or 780 mg/day or to decrease to 195 mg/day; patients can be treated for 2 years (Figure 1)

METHODS *cont.*

Figure 1. Design of the STAR 1 and STAR 2 Studies

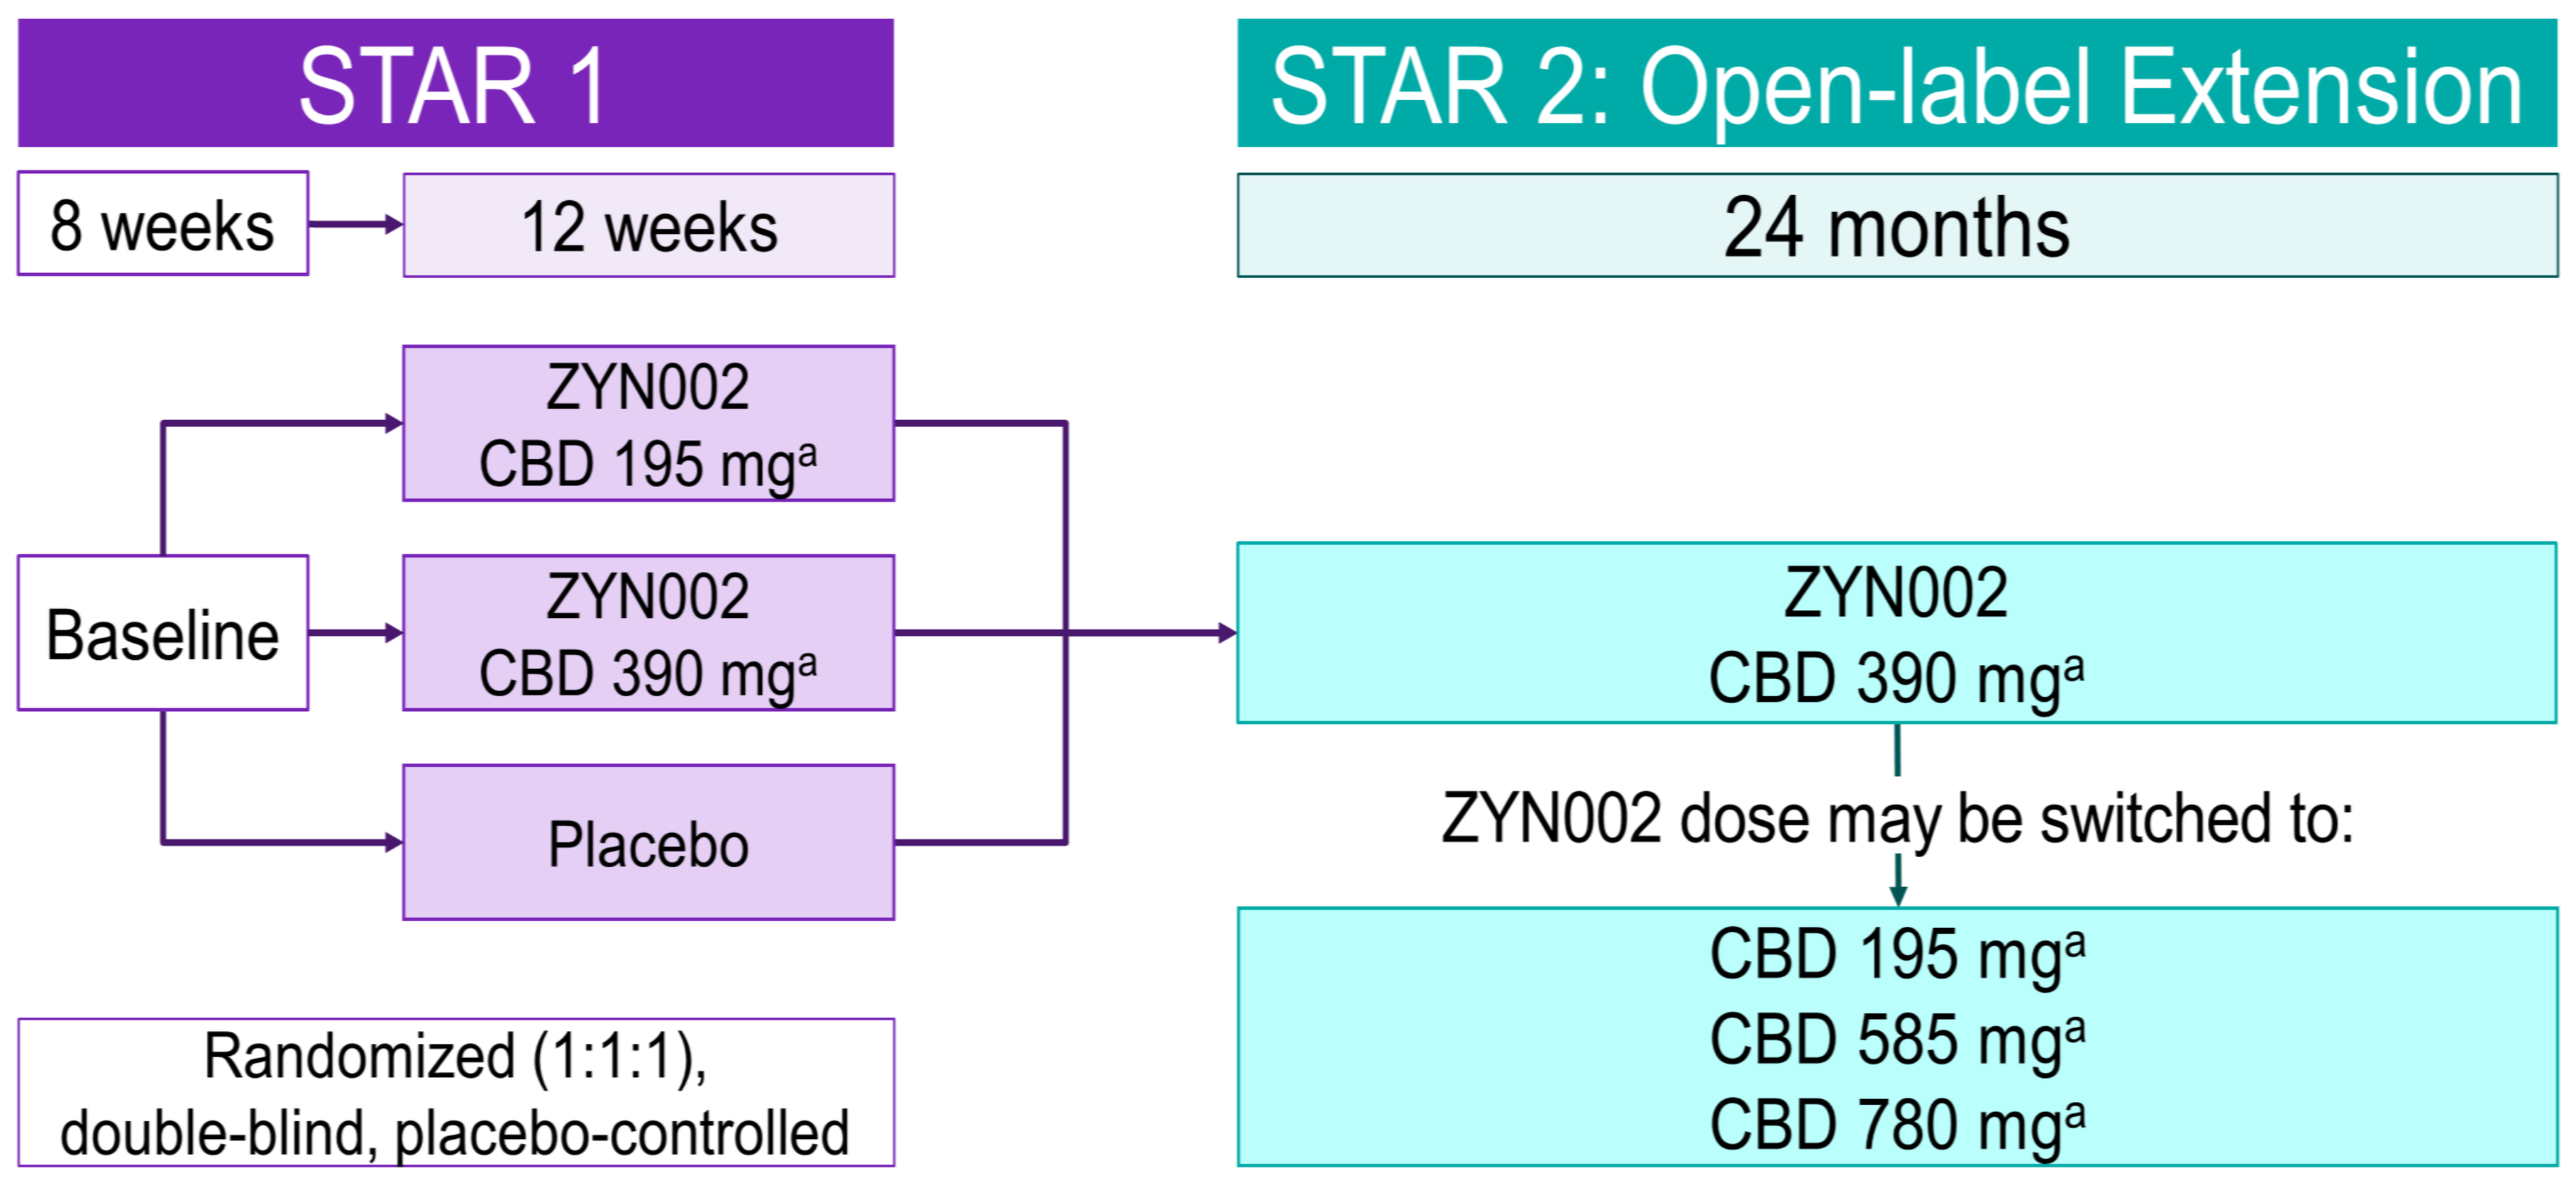
STAR 1
8 weeks
12 weeks
Baseline
ZYN002
CBD 195 mg[a]
ZYN002
CBD 390 mg[a]
Placebo
Randomized (1:1:1),
double-blind, placebo-controlled
STAR 2: Open-label Extension
24 months
ZYN002
CBD 390 mg[a]
ZYN002 dose may be switched to:
CBD 195 mg[a]
CBD 585 mg[a]
CBD 780 mg[a]

[a]Daily dose, split BID in 4.2% gel

METHODS *cont.*

Patients

- Inclusion Criteria: Aged 18 to 70 years and in generally good health at screening and at least a 2-year history of epilepsy with partial onset (focal) seizures with or without secondary generalization[3]
- All patients had their seizure history and diagnosis reviewed and confirmed by the Epilepsy Study Consortium prior to randomization
- Exclusion Criteria: Use of cannabis, CBD-, or THC-containing products within 4 weeks of screening or anytime during the study; use of any of the following AEDs: clobazam, ethosuximide, felbamate, or vigabatrin

Assessments

- STAR 1 primary efficacy analysis: Log-transformed seizure frequency per 28-day period (SF28) during the 12-week treatment period, controlling for the baseline period
- STAR 2 primary efficacy analysis: Percent reduction from STAR 1 baseline, by originally randomized group, at Months 3, 6, 9, 12, and 15 of STAR 2 (as of 12 December 2017)
- Safety: Clinical labs, physical examination, and adverse events (AEs) as of 01 February 2018

RESULTS

- 188 patients were randomized, 186 were analyzed for efficacy, and 174 completed STAR 1
- 171 patients (98% of STAR 1 completers) continued into STAR 2
- As of 12 December 2017, 95 patients remained in the STAR 2 study

Demographics and AEDs

Table 1. STAR 2 Demographic Characteristics

	Placebo	ZYN002 195 mg	ZYN002 390 mg
	n=60	n=56	n=55
Age, years			
Mean	40.4	36.7	40.3
Min, Max	18, 71	18, 64	19, 67
Male, n (%)	27 (45.0)	29 (51.8)	25 (45.5)
Female, n (%)	33 (55.0)	27 (48.2)	30 (54.5)

- Patients were taking a wide range of antiepileptic drugs (AEDs), and the most common AEDs used are: levetiracetam, carbamazepine, lamotrigine, lacosamide, and valproate

Efficacy

- By the end of STAR 1, patients who received ZYN002 195 mg or 390 mg CBD daily did not have statistically significant reductions in focal seizures versus placebo
- In STAR 2, longer exposure to ZYN002 resulted in greater improvements in seizure frequency among all ZYN002-treated patients (Figure 2), including when examined by originally randomized ZYN002 dose in STAR 1 (Figure 3)

Safety

- ZYN002 was well tolerated, with good skin tolerability, in both STAR 1 and STAR 2
- In STAR 1, there were 15 serious AEs reported; 3 occurred during the 8-week baseline period, and 1 was reported after a patient discontinued from the study

Figure 2. Median Change (%) in Seizure Rates at Months 3, 6, 9, and 12

All ZYN002-treated Patients in STAR 2

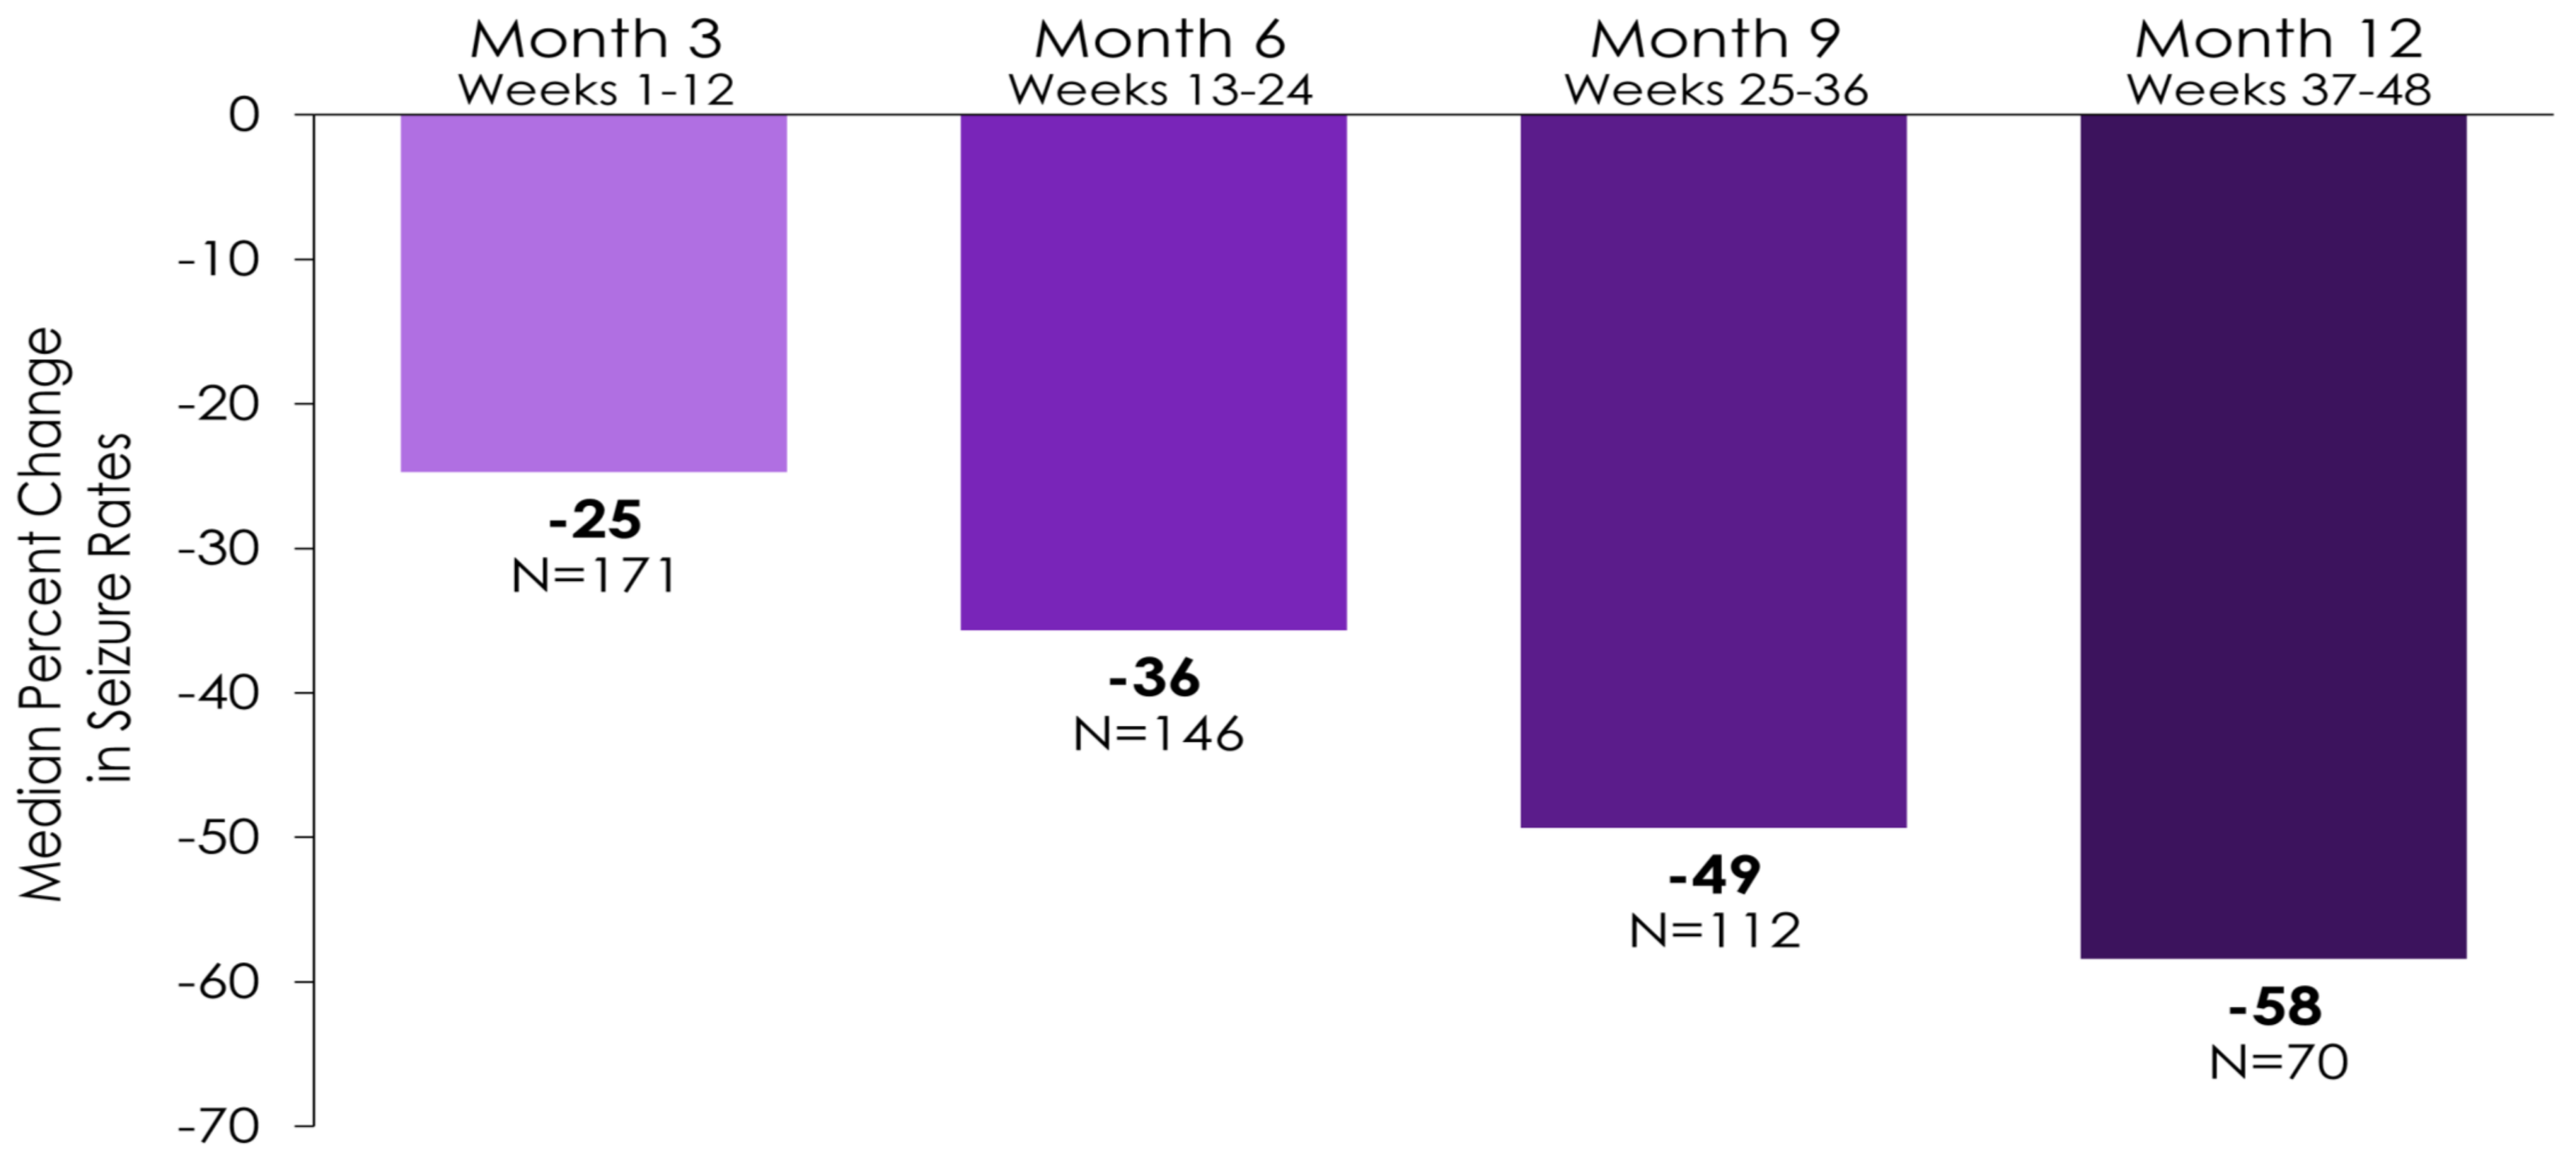
Month 3
Weeks 1-12
Month 6
Weeks 13-24
Month 9
Weeks 25-36
Month 12
Weeks 37-48
0
-10
-20
-30
-40
-50
-60
-70
Median Percent Change in Seizure Rates
-25
N=171
-36
N=146
-49
N=112
-58
N=70

Figure 3. Median Change (%) in Seizure Rates at Months 3, 6, 9, and 12

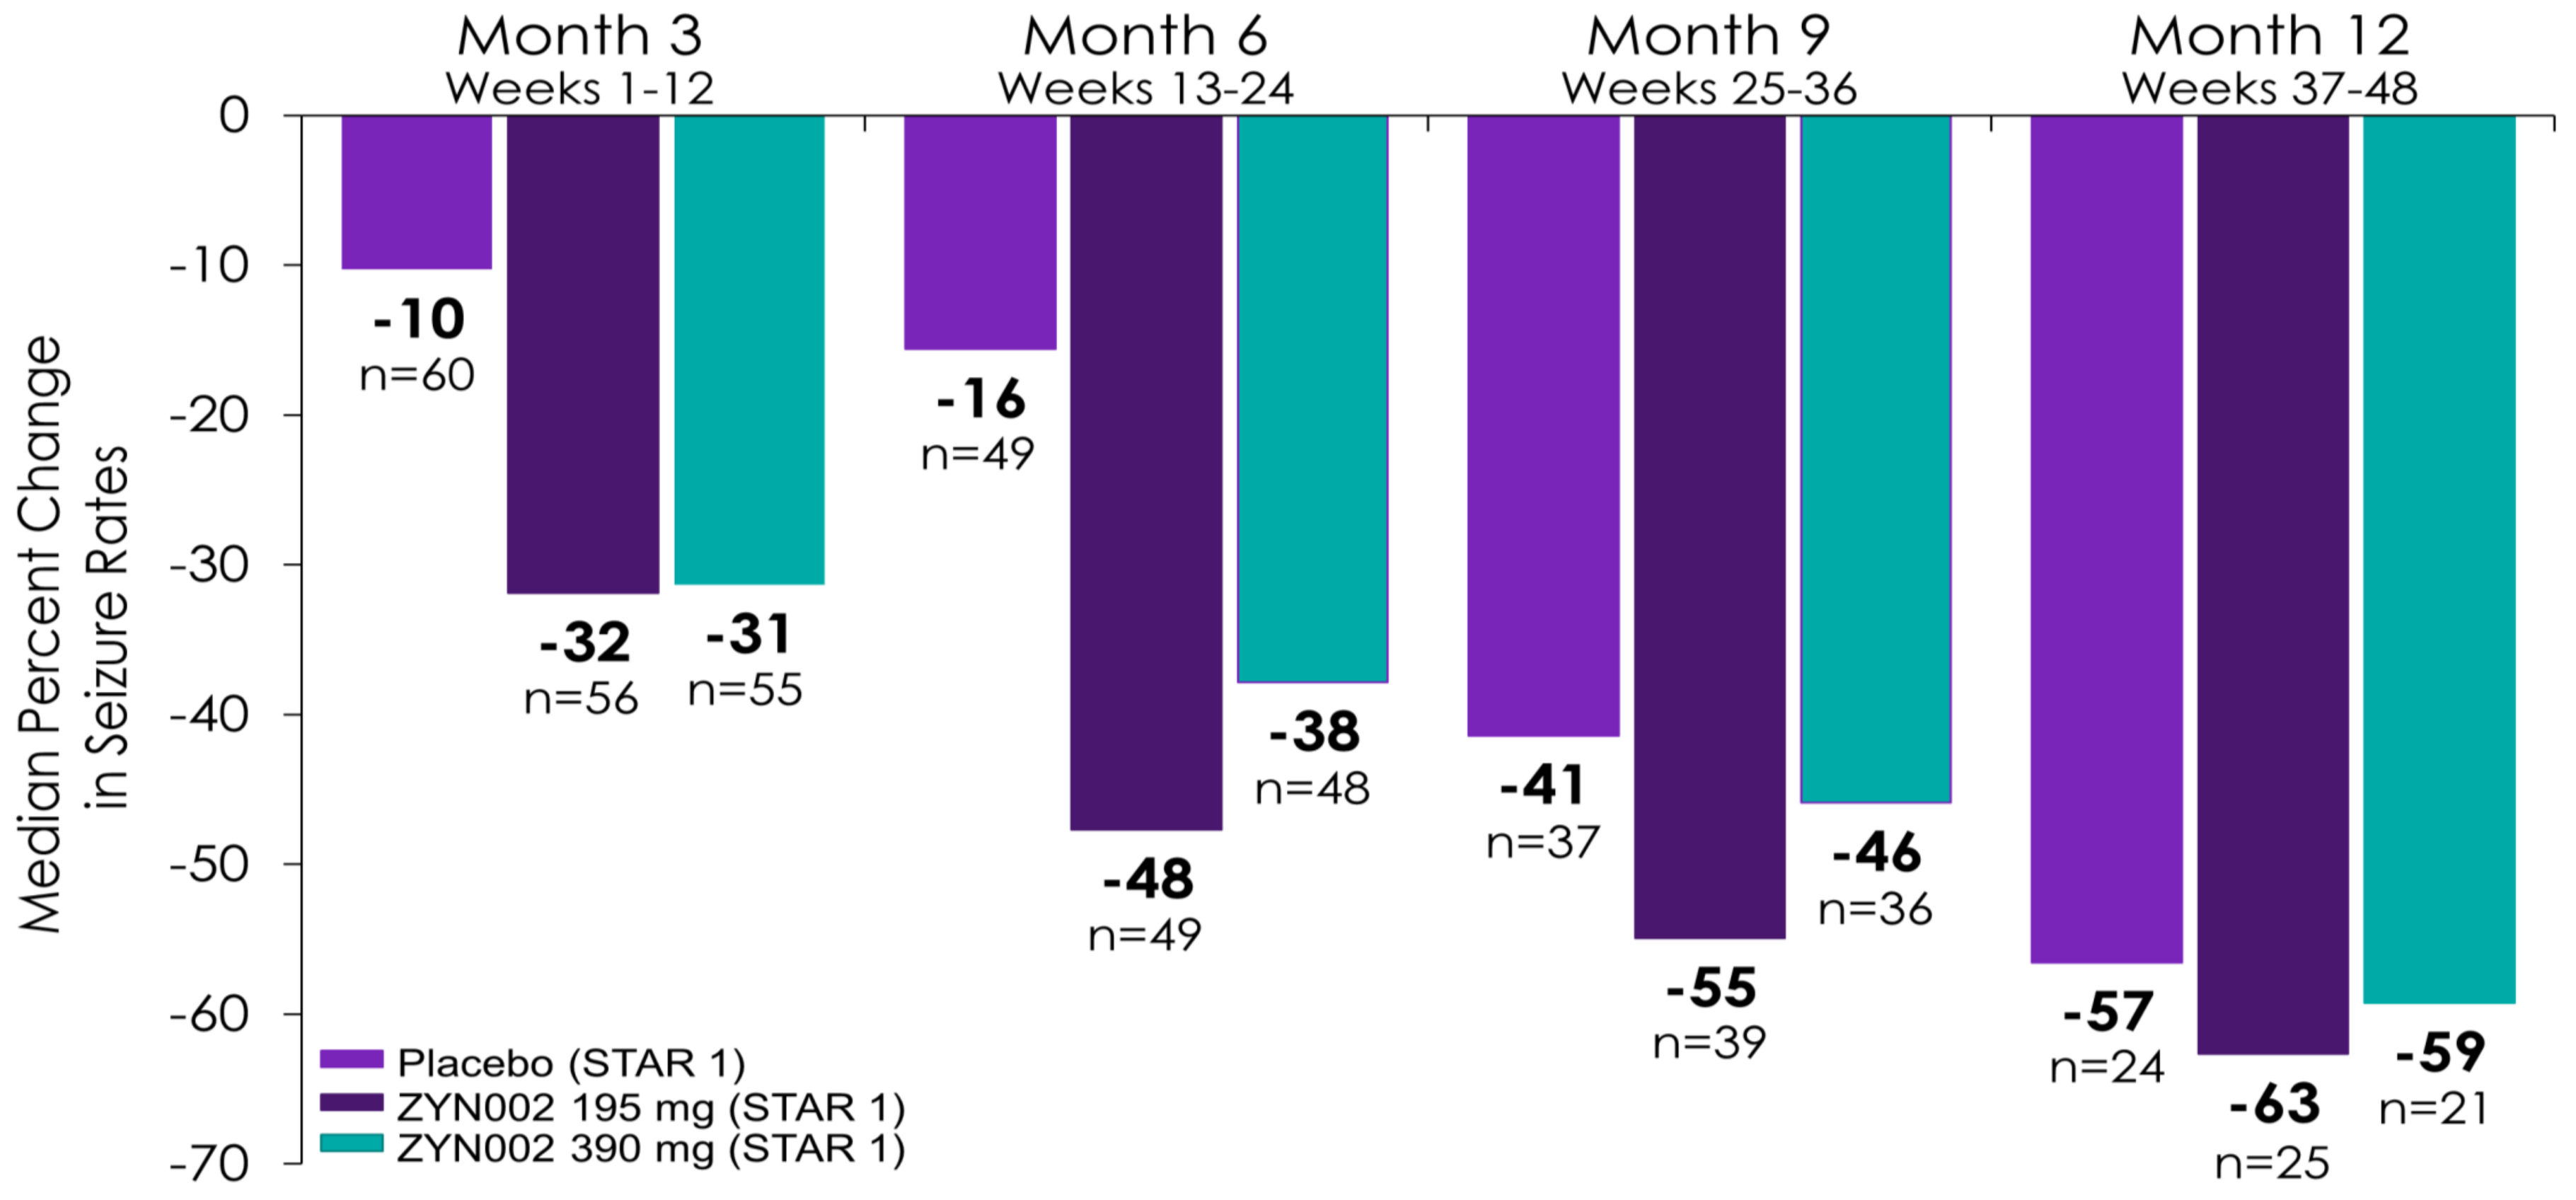
Month 3
Weeks 1-12
Month 6
Weeks 13-24
Month 9
Weeks 25-36
Month 12
Weeks 37-48
0
-10
-20
-30
-40
-50
-60
-70
Median Percent Change in Seizure Rates
-10
n=60
-32
n=56
-31
n=55
-16
n=49
-48
n=49
-38
n=48
-41
n=37
-55
n=39
-46
n=36
-57
n=24
-63
n=25
-59
n=21
Placebo (STAR 1)
ZYN002 195 mg (STAR 1)
ZYN002 390 mg (STAR 1)

RESULTS *cont.*

Safety *cont.*

- The most common AEs in STAR 1 ZYN002-treated patients were upper respiratory tract infection (viral and bacterial [16%]), headache (11%), fatigue (7%), and laceration (5%)
- No serious AE was considered related to study drug in STAR 1
- In STAR 2, the most common treatment-emergent AEs (>7.5%) were upper respiratory tract infection (viral and bacterial [15%]) and headache (11%)
- One serious AE was considered possibly related to ZYN002 (increased anxiety) in STAR 2

CONCLUSIONS

- In STAR 1, the change in seizure frequency did not statistically differ between placebo and both doses of ZYN002
- The unblinded use of ZYN002 for an additional 12 months during STAR 2 resulted in clinically meaningful seizure reductions both across and within the originally randomized STAR 1 groups
- In STAR 2, there were too few patients at 585 mg and 780 mg doses at this time to report individual differences as a function of dose
- ZYN002 was well tolerated, and there were no abnormal liver AEs (ie, alanine aminotransferase/aspartate aminotransferase > 3x upper limit of normal)
- These data indicate further study with longer-term blinded evaluation is warranted

REFERENCES

1. Mechoulam R et al. *Chem Phys Lipids*. 2002;121(1-2):35-43.
2. Gloss D et al. *Cochrane Database Syst Rev*. 2014(3):Cd009270.
3. Fisher RS. *Curr Neurol Neurosci Rep*. 2017;17(6):48.

Presented at the 2018 Annual Meeting of the American Academy of Neurology (P4.468), April 21-27, 2018, Los Angeles, CA

JM is a paid consultant of Zynerba; TOB, SFB, and JF have received honoraria and research support from Zynerba; MBM and DG are employees of Zynerba Pharmaceuticals